INCREASED TEAR FILM LIPID LAYER THICKNESS WITH THE USE OF TWO NOVEL LUBRICANT EYE DROPS

Donald R. Korb,[1] Robert C. Scaffidi,[2] Jack V. Greiner,[2,3] Kenneth R. Kenyon,[2,3] John P. Herman,[4] Caroline A. Blackie,[1] Thomas Glonek,[5] Courtney L. Case,[1] Victor M. Finnemore,[1] Teresa Douglass[1]

[1]Korb Associates, Boston, MA; [2]Schepens Eye Research Institute, Boston, MA; [3]Department of Ophthalmology, Harvard Medical School, Boston, MA; [4]Pittsfield Eye Associates, Pittsfield, MA; [5]Midwestern University, Downers Grove, IL

ABSTRACT

Purpose: Tear film lipid layer thickness (LLT) has been correlated to the presence of dry eye symptoms. Two novel products, Soothe™ and Systane™, have been introduced for the lubrication of dry eye and relief of symptoms. This study was conducted to determine if a single eye drop of either product produced a significant increase in LLT for subjects reporting symptoms indicative of dry eyes.

Methods: A double-blind, internally-paired study was performed. A custom lipid layer interferometer, enabling characterization of lipid layer interference patterns, was used to quantify LLT (OU) of eligible subjects. Subjects (n=40) received a single drop of Soothe™ in one eye and a single drop of Systane™ in the other eye. Test drops were supplied in masked identical opaque eye dropper bottles. The LLT of all subjects, following the instillation of each test drop, was analyzed. Inclusion criteria included: 1) presence of dry eye symptoms and 2) LLT < 75 nm, with variability no greater than ± 15 nm in each eye or between an individual pair of eyes, over a 15 minute observation period.

Results: The mean ± standard error baseline LLT findings pre-eye drop instillation were 60.0 ± 1.8 nm for eyes treated with Soothe™, and 61.5 ± 1.8 nm for eyes treated with Systane™. These means were not significantly different (p > 0.5). Post-eye drop instillation, the mean LLT for eyes treated with Soothe™ more than doubled from 60.0 nm to 144.4 ± 4.7 nm, and the mean LLT for eyes treated with Systane™ increased from 61.5 nm to 84.3 ± 4.9 nm. The mean increase in LLT for Soothe™ was significantly greater than that produced by the weakest measurable blink response (p < 0.0001). The mean increase in LLT for Systane™ was not significantly greater than that produced by the weakest measurable blink response (p = 0.07).

Conclusions: In subjects with symptoms indicative of dry eye states and with LLT ≤ 75 nm, one eye drop of Soothe™ resulted in a significant increase in LLT, more than doubling LLT, while the increase for Systane™ was not significantly greater than that produced by the weakest measurable blink response.

INTRODUCTION

Dry eye is a commonly reported condition in clinical practice.[1] According to the National Eye Institute/Industry Workshop on clinical trials in dry eyes, most forms of dry eye exhibit the following four features: 1) symptoms; 2) interpalpebral surface damage; 3) tear (film) instability; and 4) tear hyperosmolarity.[1]

www.sootheeyedrops.com 1

One of these features, tear film stability, has been noted to be dependent upon the thickness of the lipid layer[2-5] Lipid layer thickness (LLT) has been correlated to other dry eye tests signifying that the measurement of LLT is a reliable test for the diagnosis of dry eye.[6]

Standard dry eye treatments, i.e. lubricant eye drops, offer little or no relief, and may even increase tear evaporation rates[7,8] These studies have accelerated the pursuit of a more effective lubricant eye drop. This study examines two new lubricant eye drops: Soothe™ (Alimera Sciences, Inc., Alpharetta, GA) and Systane™ (Alcon Laboratories, Inc., Fort Worth, TX). Both eye drops are marketed to relieve the symptoms of dry eye.

Purpose: To assess and compare the effects of Soothe™ and Systane™ on the tear film LLT of subjects experiencing dry eye, and to quantify any change in LLT due to either product.

METHODS

Subjects (n = 40, median age = 40.3, range 23-69 years) were enrolled following an ocular examination, medical history screening, and baseline LLT evaluation.

Inclusion Criteria:

- Dry eye symptoms determined by questionnaire[9]
- LLT ≤ 75 nm (Baseline)
- LLT fluctuations ≤ 15 nm over a period of 15 min
- Difference in LLT between right & left eyes ≤ 15 nm
- Increase in LLT after 3 forceful blinks ≤ 15 nm
- Monocular BCVA ≥ 20/70
- Unremarkable biomicroscope findings

Exclusion Criteria:

- Use of any eye lubricant or ointment during the 36 hours prior to the study
- Use of contact lenses or excessive eye cosmetics on the day of the study
- Presence of ocular/systemic disease influencing the tear film
- Anisometropia ≥ 3.00 D or astigmatism ≥ 2.75 D in either eye

Table 1: Quantification of lipid layer thickness (LLT) according to the dominant color(s) in the interference pattern.

Color	LLT (nm)
Gray/White	45
Gray	60
Gray/Yellow	75
Yellow	90
Yellow/Brown	105
Brown/Yellow	120
Brown	135
Brown/Blue	150
Blue/Brown	165
Blue	180

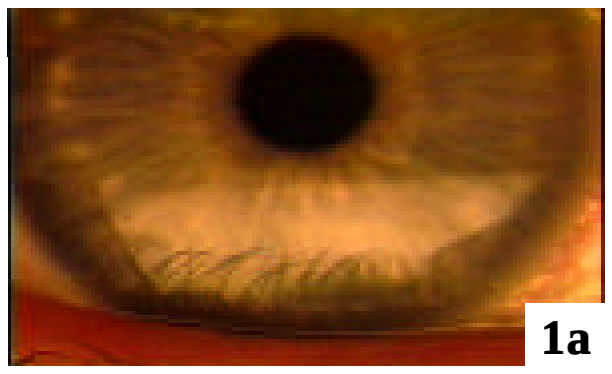
1a

Baseline - Poor

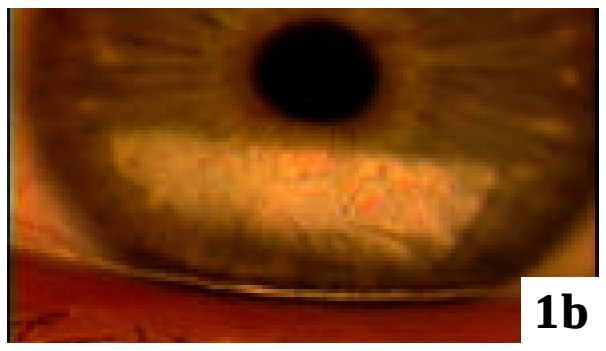
1b

Soothe™ 1 Minute - Excellent

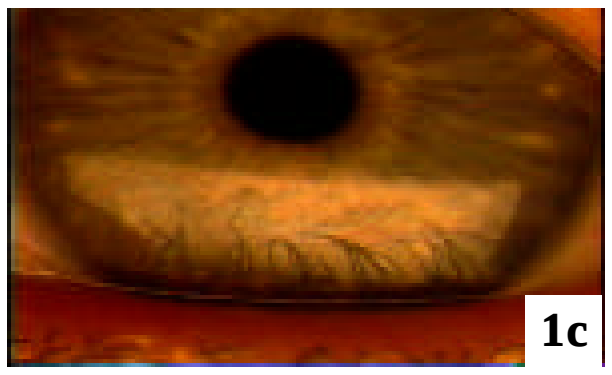
1c

Soothe™ 5 Minutes - Excellent

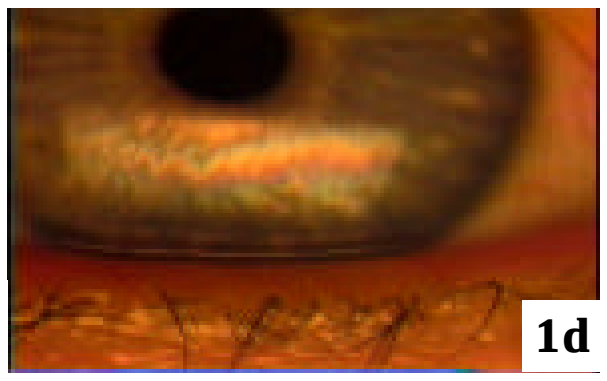
1d

Soothe™ 15 Minutes - Excellent

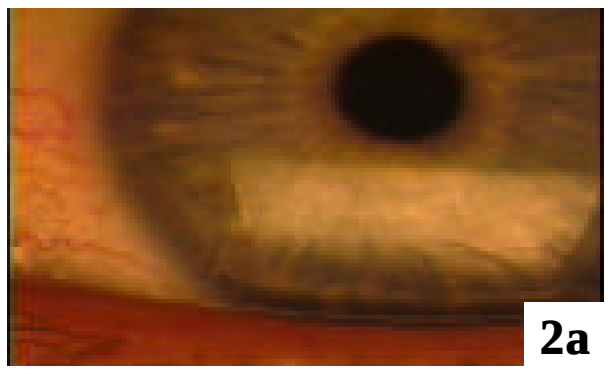
2a

Baseline - Poor

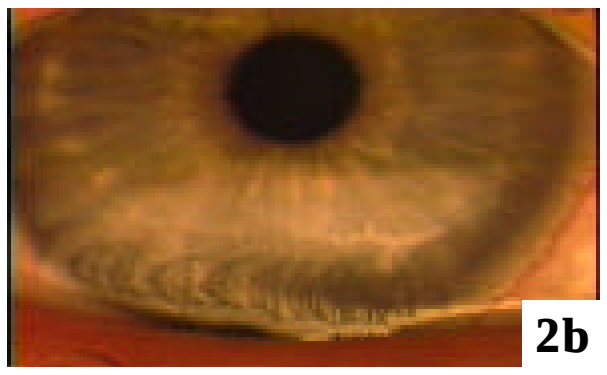
2b

Systane™ 1 Minute - Poor

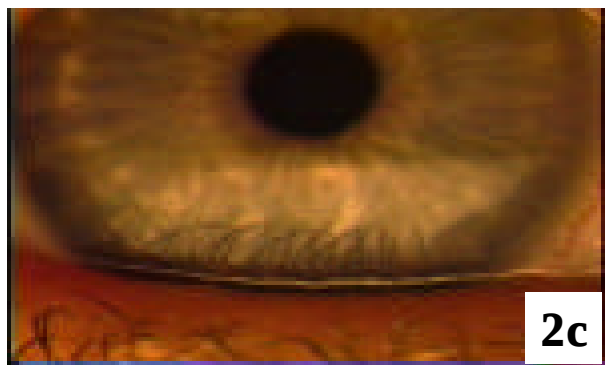
2c

Systane™ 5 Minutes - Poor

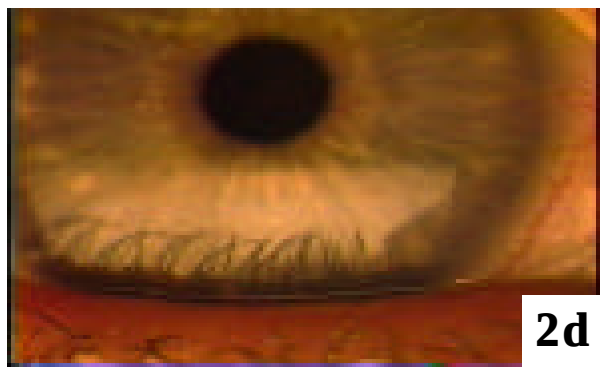
2d

Systane™ 15 Minutes - Poor

www.sootheeyedrops.com 2

- Any corneal, conjunctival, or eyelid abnormalities
- Known sensitivity to any of the ingredients contained in Soothe™ or Systane™

Of the 49 subjects excluded from this study, 49.0% (n=24) were excluded solely because forceful blinking increased LLT more than 15 nm. In addition, 28.6% (n=14) were excluded for multiple reasons, one of which was related to forceful blinking.

The LLT of each eye was observed using a custom-designed lipid layer interferometer.[10] The LLT was evaluated at time 0 min, and again at 15 min to assure a consistent baseline LLT. The temperature and relative humidity of the examining room were monitored and remained constant at 25°C and 35-50% relative humidity. Test drops were supplied in identical, masked opaque 3 mL dropper bottles. Subjects received a single drop of Soothe™ in one eye and a single drop of Systane™ in the other eye. The order of test drop and the eye in which it was instilled were randomized. The test drops were instilled into the conjunctival sac of each eye at the appropriate test time (each eye drop instillation was timed separately). The subject was then instructed to blink normally while fixating on a target and to abstain from rubbing the eyes or face. If staring persisted for >10 sec, the subject was instructed to blink normally. Thereafter, the LLT was measured at 1 min, 5 min, and 15 min post-eye drop instillation. All observations of LLT were recorded using a videotape.[10] The LLT was later quantified by two independent and trained observers using a published grading scheme (**Table 1**).[6,10]
dry eye disorders, probably because instrumentation is not readily available.

RESULTS

A paired t-test was used to determine the statistical significance of any measured effect on the LLT of Soothe™ relative to Systane™. This is the statistical test of choice given the internally paired nature of the data. By directly comparing LLT change in right and left eyes of the same individual, genetic and environmental differences between subjects were effectively controlled.

The mean ± standard error baseline LLT findings pre-eye drop instillation were 60.0 ± 1.8 nm for eyes treated with Soothe™, and 61.5 ± 1.8 nm for eyes treated with Systane™. These means were not significantly different ($p > 0.5$).

Post-eye drop instillation, the mean LLT for eyes treated with Soothe™ increased 84.4 to 144.4 ± 4.7 nm (**Fig 1 a-d**), and the mean LLT for eyes treated with Systane™ increased 28.8 to 84.3 ± 4.9 nm (**Fig 2 a-d**). The measured LLT post-eye drop instillation was the thickest value of LLT achieved regardless of the time point at which it occurred. The LLT increase from Soothe™ was significantly greater than that from Systane™ ($p < 0.0001$).

The weakest (smallest) measurable blink response, as measured with the lipid layer interferometer, is an increase in LLT of 15 nm secondary to a blink. When utilizing the weakest measurable blink response of 15 nm as a control, the LLT increase secondary to Soothe™ was highly significant ($p > 0.0001$) but Systane™ produced no significant increase in the LLT ($p = 0.07$) (t-test of individual means).

DISCUSSION

The lipid layer is recognized as a critical component of both tear film stability and ocular comfort.[2-5] However, lipid layer evaluation has not been routinely employed for the diagnosis or treatment of dry eye disorders , probably because instrumentation is not readily available.

www.sootheeyedrops.com 3

These studies were conducted in an examining room with 35-50% humidity. Further studies to evaluate the two products in lower humidity environments would broaden the knowledge of the function of these two products under more adverse conditions, and would ideally be conducted in a controlled adverse environment.[11-13]

In this study, particular emphasis was directed to controlling the variables encountered in LLT evaluation, but not usually recognized.[10,14-16] These included recognizing that LLT and lipid layer characteristics are a function of the nature and frequency of the blink,[10] and the dynamic rather than static design of the lipid layer.[10,15]

The lipid layer is usually thickest slightly above the meniscus, and gradually thins as the result of the tear film and lipid layer being pulled upward over the corneal surface upon the return aspect of the blink. **Thus, it is important to evaluate the lipid layer in a defined area.**

Figure 1: Color Interference Patterns of Eye Treated with Soothe™

a: LLT prior to Soothe™: Dominant colors = gray/white
LLT = 45 nm (Classification = **POOR**)
b: LLT 1 minute following Soothe™: Dominant colors = brown/blue
LLT = 150 nm (Classification = **EXCELLENT**)
c: LLT 5 minutes following Soothe™: Dominant colors = blue/brown
LLT = 165 nm (Classification = **EXCELLENT**)
d: LLT 15 minutes following Soothe™: Dominant colors = blue/brown
LLT = 165 nm (Classification = **EXCELLENT**)

Figure 2: Color Interference Patterns of Eye Treated with Systane™

a: LLT prior to Systane™: Dominant colors = gray/white
LLT = 45 nm (Classification = **POOR**)
b: LLT 1 minute following Systane™: Dominant colors = gray/white
LLT = 45 nm (Classification = **POOR**)
c: LLT 5 minutes following Systane™: Dominant colors = gray/white
LLT = 45 nm (Classification = **POOR**)
d: LLT 15 minutes following Systane™: Dominant colors = gray/white
LLT = 45 nm (Classification = **POOR**)

Observation immediately after the upward movement of the eyelid usually exhibits the colors and characteristics indicative of the maximum thickness of the individual’s lipid layer. As the interblink phase continues, the colors and characteristics of the lipid layer gradually fade and change, indicating thinning of the lipid layer. Following the next blink, the lipid layer will reform and may increase in thickness, depending upon the amount of lipid expressed from the meibomian glands and the amount of lipid available in the meniscus. Partial blinking will not express the meibomian glands or reform those portions of the lipid layer that are not wiped by the blink; the unwiped areas will continue to thin until rewiped by a subsequent blink.

Thus, it is important to observe the lipid layer over a period of 30-60 sec and 5 or more blink cycles to ensure that the measurement of LLT is consistent and representative of the typical thickness of the lipid layer.

www.sootheeyedrops.com 4

Since LLT may increase as a function of blinking,[10] subjects whose LLT increased by >15nm after forceful blinking were excluded from the study to avoid confounding the results. This ensured that any increase in LLT would be a result of the application of the test eye drop, and not the result of the blink. With these controls in place, it was found that Soothe™ was very effective in increasing LLT, the thickness more than doubling (141% mean increase). In contrast, the increase in LLT following the instillation of one drop of Systane™ was not significantly greater than that produced by the weakest measurable blink response.

CONCLUSIONS

In subjects with symptoms indicative of dry eye states and with LLT $\leq$ 75 nm, one eye drop of Sootheä resulted in a significant increase in LLT, more than doubling LLT, while the increase for Systaneä was not significantly greater than that produced by the weakest measurable blink response.

This study was supported by Alimera Sciences, Inc. and Ocular Research of Boston, Inc.

REFERENCES

- Lemp MA. Report of the National Eye Institute/Industry Workshop on clinical trials in dry eyes. *CLAO J* 1995; 21:221-232.
- Korb DR, Greiner JV. Increase in tear film lipid layer thickness following treatment of meibomian gland dysfunction. In: Sullivan DA, ed. *Lacrimal Gland, Tear Film, and Dry Eye Syndromes*, New York: Plenum Press, 1994;293-298.
- Yokoi N, Takehisa Y, Kinoshita S. Correlation of tear lipid layer interference patterns with the diagnosis and severity of dry eye. *Am J Ophthalmol* 1996;122:818-824.
- Craig JP, Tomlinson A. Importance of the lipid layer in human tear film stability and evaporation. *Optom Vis Sci* 1997;74:8-13.
- Guillon M, Styles E, Guillon JP, Maissa C. Preocular tear film characteristics of non-wearers and soft contact lens wearers. *Optom Vis Sci* 1997;74:273-279.
- Isreb MA, Greiner JV, Korb DR, Glonek T, Mody SS, Finnemore VM, Reddy CV. Correlation of lipid layer thickness measurements with fluorescein tear film break-up time and Schirmer's test. *Eye* 2003;17:79-83.
- Trees GR, Tomlinson A. Effect of artificial tear solutions and saline on tear film evaporation. *Optom Vis Sci* 1990;67:886-890.
- Calonge M. The treatment of dry eye. *Surv Ophthalmol* 2001;45:S227-S239.
- Korb DR, Herman JP, Greiner JV, Scaffidi RC, Finnemore VM, Exford JM, Blackie CA, Douglass T. Lid wiper epitheliopathy and dry eye symptoms. (in press)
- Korb DR, Baron DF, Herman JP, Finnemore VM, Exford JM, Londono Hermosa J, Leahy CD, Glonek T, Greiner JV. Tear film lipid layer thickness as a function of blinking. *Cornea* 1994;13:354-359.
- Giovanoni A, Abelson MB, Rosen S, Welch D. A controlled environment approach to the study of dry eye [ARVO Abstract] *Invest Ophthalmol Vis Sci* 1998;39(4):S537. Abstract nr 2467.
- Ousler GW, Crampton HJ, Abelson MB. An evaluation of signs and symptoms associated with the caruncle and plica semilunaris after exposure to a controlled adverse environment [ARVO Abstract]. *Invest Ophthalmol Vis Sci* 2000;41(4):S928. Abstract nr 4936.
- Ousler GW, Abelson MB, Nally LA, Welch D, Casavant JS. Evaluation of the time to "natural compensation" in normal and dry eye subject populations during exposure to a controlled adverse environment. In: Sullivan DA, ed. *Lacrimal Gland, Tear Film, and Dry Eye Syndromes* 3 Part B, New York: Kluwer Academic/Plenum Publishers, 2002;1057-63.
- Korb DR, Greiner JV, Glonek T, Esbah R, Finnemore VM, Whalen AC. Effect of periocular humidity on the tear film lipid layer. *Cornea* 1996;15:129-134.
- Korb DR. The tear film – its role today and in the future. In: Korb DR, ed. *The Tear* Film, Oxford: Butterworth Heinemann, 2002:154-164.
- Guillon, JP. The tear film structure of the contact lens wearer. Ph.D. thesis, The City University Department of Optometry and Visual Science, 1987.

www.sootheeyedrops.com 5